#374516: Development of a key performance indicator (KPI) for real-time monitoring of treatment of early breast cancer to control sociodemographic and Indigenous system bias

Authors: Euan Walpole, Philippa Youl, Danica Cossio, Michelle Morris, Shoni Philpot. Cancer Alliance Queensland, Brisbane, Australia

Introduction
- Disparities in outcomes for some population groups continue to exist for women with breast cancer.
- Identifying optimal time to complete treatment may help reduce disparities.

Methods
- Retrospective population-based study of 8,279 women with stage I-III breast cancer who received surgery, chemotherapy (CT) and radiation therapy (RT).
- Calculated time from diagnosis to completing all treatments.
- Using Stata's empirical cut-point a threshold of 37 weeks to complete treatment was identified. Optimal time to start each treatment modality was also identified.
- Cox proportional hazards was used to examine factors associated with risk of death and included time to each treatment as a covariate.

*Implementation of a KPI for delay in receiving adjuvant chemotherapy of **6.9 weeks** can provide a real-time target for patient care*

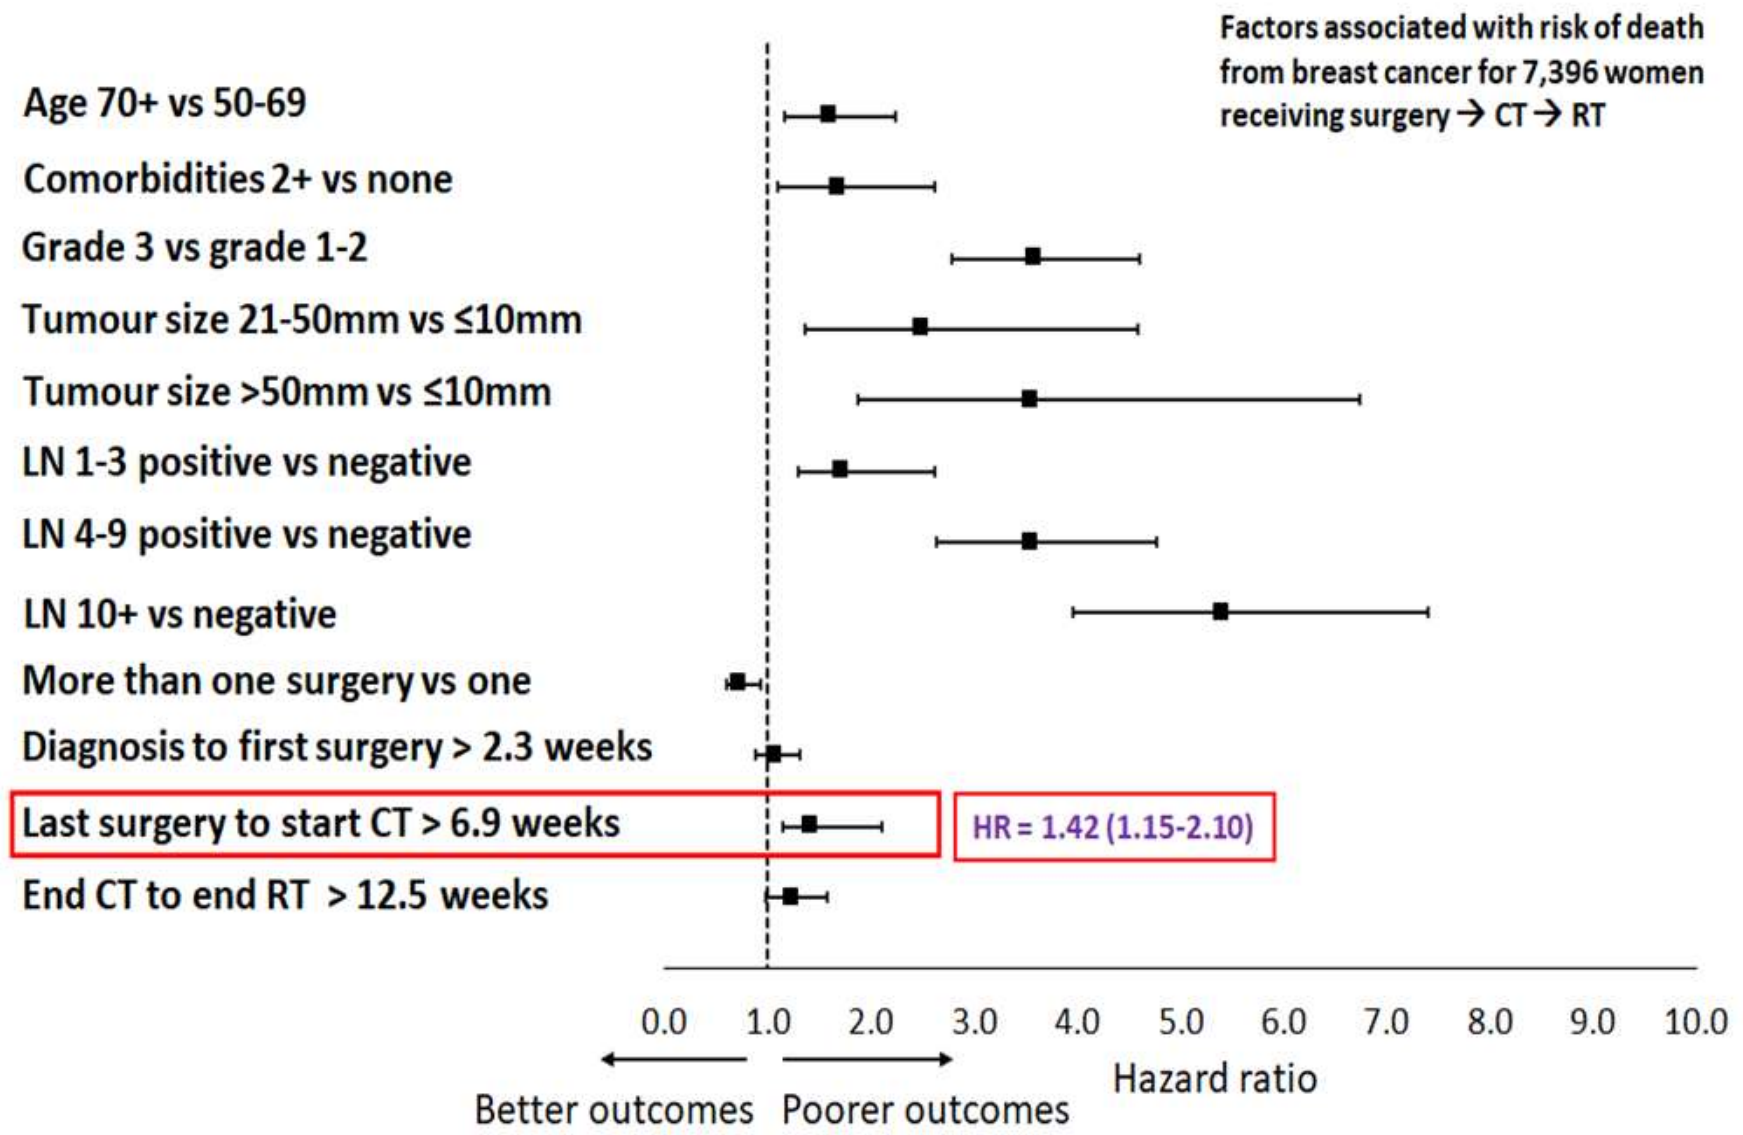
Factors associated with risk of death from breast cancer for 7,396 women receiving surgery → CT → RT
Age 70+ vs 50-69
Comorbidities 2+ vs none
Grade 3 vs grade 1-2
Tumour size 21-50mm vs ≤10mm
Tumour size >50mm vs ≤10mm
LN 1-3 positive vs negative
LN 4-9 positive vs negative
LN 10+ vs negative
More than one surgery vs one
Diagnosis to first surgery > 2.3 weeks
Last surgery to start CT > 6.9 weeks
HR = 1.42 (1.15-2.10)
End CT to end RT > 12.5 weeks
0.0 1.0 2.0 3.0 4.0 5.0 6.0 7.0 8.0 9.0 10.0
Hazard ratio
Better outcomes
Poorer outcomes

Results
- Overall survival (OS) was 94% at 5 years for treatment completion < 37 weeks and 89% for ≥ 37 weeks (p <0.001).
- Being Indigenous (p=0.002), disadvantaged (p=0.003) and ≥two treatment modalities in public sector (p <0.001) were associated with likelihood of completing treatment ≥37 weeks.
- Risk of death increased by **42%** when time from surgery to starting CT was beyond 6.9 weeks.
- When delay from surgery to CT used in multivariate analysis, Indigenous and disadvantage are no longer significant.
- KPIs can be monitored using funnel plot help identify treatment facilities falling outside recommended treatment times.

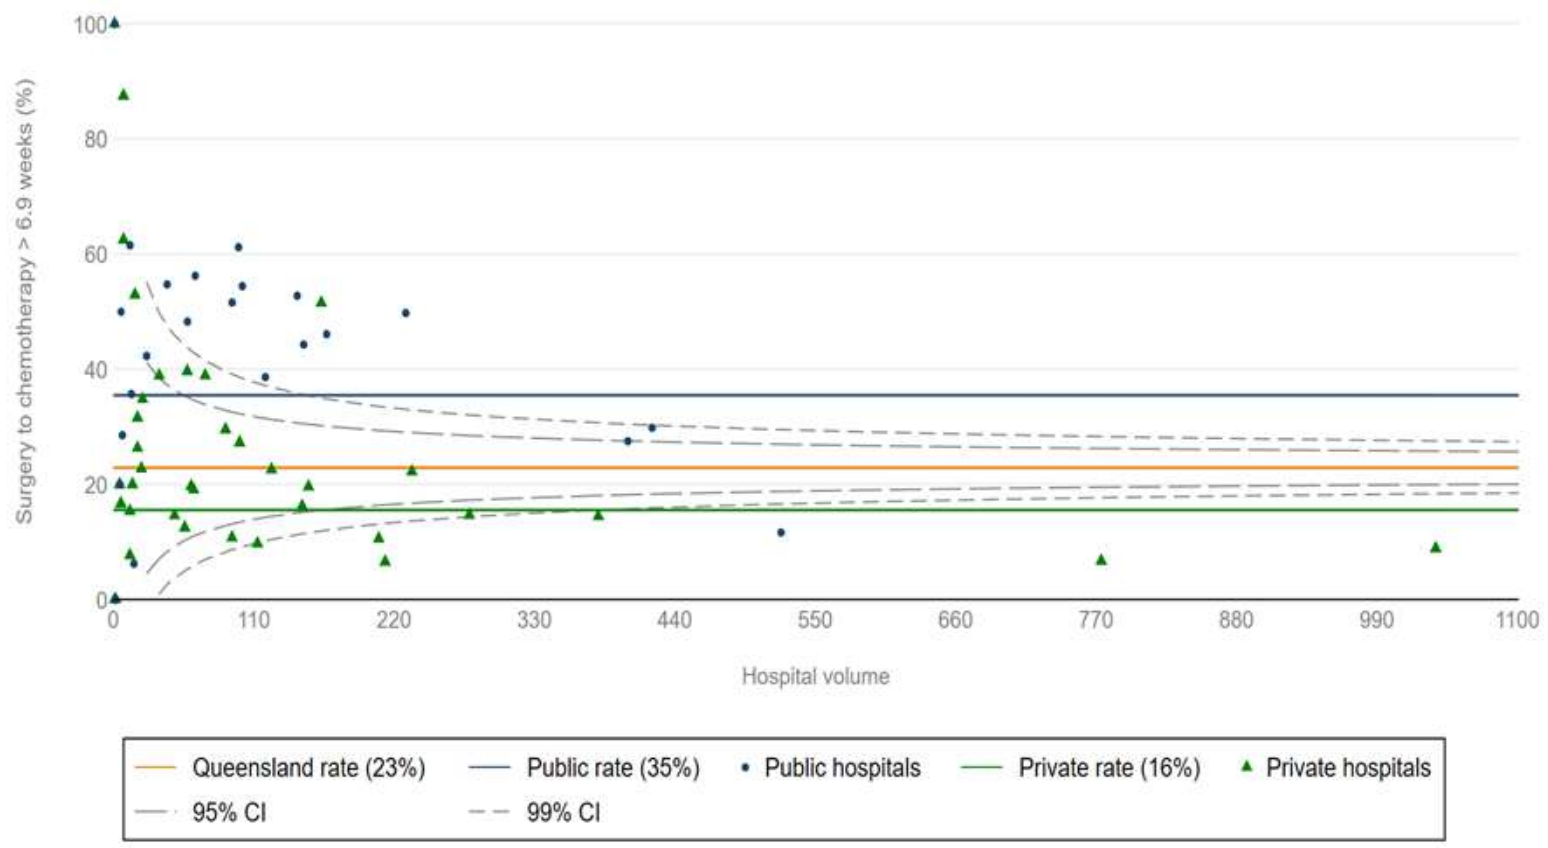
Surgery to chemotherapy > 6.9 weeks (%)
100
80
60
40
20
0
0 110 220 330 440 550 660 770 880 990 1100
Hospital volume
Queensland rate (23%)
Public rate (35%)
Public hospitals
Private rate (16%)
Private hospitals
95% CI
99% CI

Where to from here?
Systematic reporting of this KPI has potential to remove treatment biases for Indigenous and lower socioeconomic patients.

cancer alliance queensland

the Partnership queensland cancer control safety and quality partnership

qccat queensland cancer control analysis team

qcr queensland cancer register